133TiP

A Randomized Phase 3 Study of Ivonescimab Versus Pembrolizumab as First-Line Treatment of Metastatic Non–Small Cell Lung Cancer With High Tumor PD-L1 Expression: HARMONi-7

Antonio Passaro,[1] Nicolas Girard,[2] Delvys Rodriguez-Abreu,[3] Mahmut Gumus,[4] Yi-Long Wu,[5] Konstantinos Syrigos,[6] Rodryg Ramlau,[7] Edgardo S. Santos,[8] Chintan Gandhi,[9] Natalie Vokes,[10] Bo Wang,[11] Julia Rotow,[12] Paul Paik,[13] Jared Weiss,[14] Jason Agulnik,[15] Xiang Bai,[16] Patti McGovern,[16] Antonio Calles[17]

[1]European Institute of Oncology, Milan, Italy; [2]Institut Curie, Paris, France; [3]Universidad de Las Palmas de Gran Canaria, Las Palmas de Gran Canaria, Spain; [4]Istanbul Medeniyet University, Istanbul, Turkey; [5]Guangdong Lung Cancer Institute, Guangzhou, China; [6]Athens University School of Medicine, Athens, Greece; [7]Poznan University of Medical Sciences, Poznan, Poland; [8]The Oncology Institute of Hope and Innovation, Fort Lauderdale, FL, USA; [9]South Florida Oncology & Hematology Consultants, Tamarac, FL, USA; [10]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [11]Willamette Valley Cancer Institute and Research Center, Eugene, OR, USA; [12]Dana-Farber Cancer Institute, Boston, MA, USA; [13]Thoracic Oncology Service, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [14]UNC Lineberger Comprehensive Cancer Center, Chapel Hill, NC, USA; [15]Jewish General Hospital, Montreal, QC, Canada; [16]Summit Therapeutics, Menlo Park, CA, USA; [17]Hospital General Universitario Gregorio Marañón, Madrid, Spain

BACKGROUND

- Anti–programmed cell death protein 1 (PD-1) and programmed cell death ligand 1 (PD-L1) as monotherapies or in combination with chemotherapy are the standard of care for first-line treatment of PD-L1–positive locally advanced and metastatic non–small cell lung cancer (mNSCLC) in the absence of activating mutations[1]
- The addition of anti-angiogenic agents to standard first-line treatment with a PD-L1 inhibitor and platinum-based chemotherapy has shown modest efficacy in patients with nonsquamous mNSCLC[2]
- Ivonescimab, a first-in-class investigational bispecific antibody against PD-1 and vascular endothelial growth factor, showed promising clinical efficacy in the HARMONi-A and HARMONi-2 phase 3 trials in China[3,4]
- In HARMONi-2, progression-free survival (PFS) improved significantly in patients with PD-L1–positive (tumor proportion score [TPS] ≥1%) advanced NSCLC with ivonescimab compared with pembrolizumab (median PFS, 11.1 and 5.8 months, respectively; hazard ratio, 0.51; one-sided $P < 0.0001$)[4]
 - Results of the trial also showed an acceptable safety profile for ivonescimab, including in patients with squamous and nonsquamous NSCLC

TRIAL DESIGN

- HARMONi-7 (NCT06767514) is a global, randomized, double-blind, phase 3 study to compare the efficacy and safety of ivonescimab against pembrolizumab monotherapy in patients with stage IV mNSCLC with high tumor PD-L1 expression and no known actionable genomic alterations who have not previously received systemic therapy for metastatic disease (**Figure 1**)
- Approximately 780 patients will be randomly assigned (1:1) to receive ivonescimab 20 mg/kg or pembrolizumab 200 mg every 3 weeks for up to 24 months

Figure 1. Trial Design

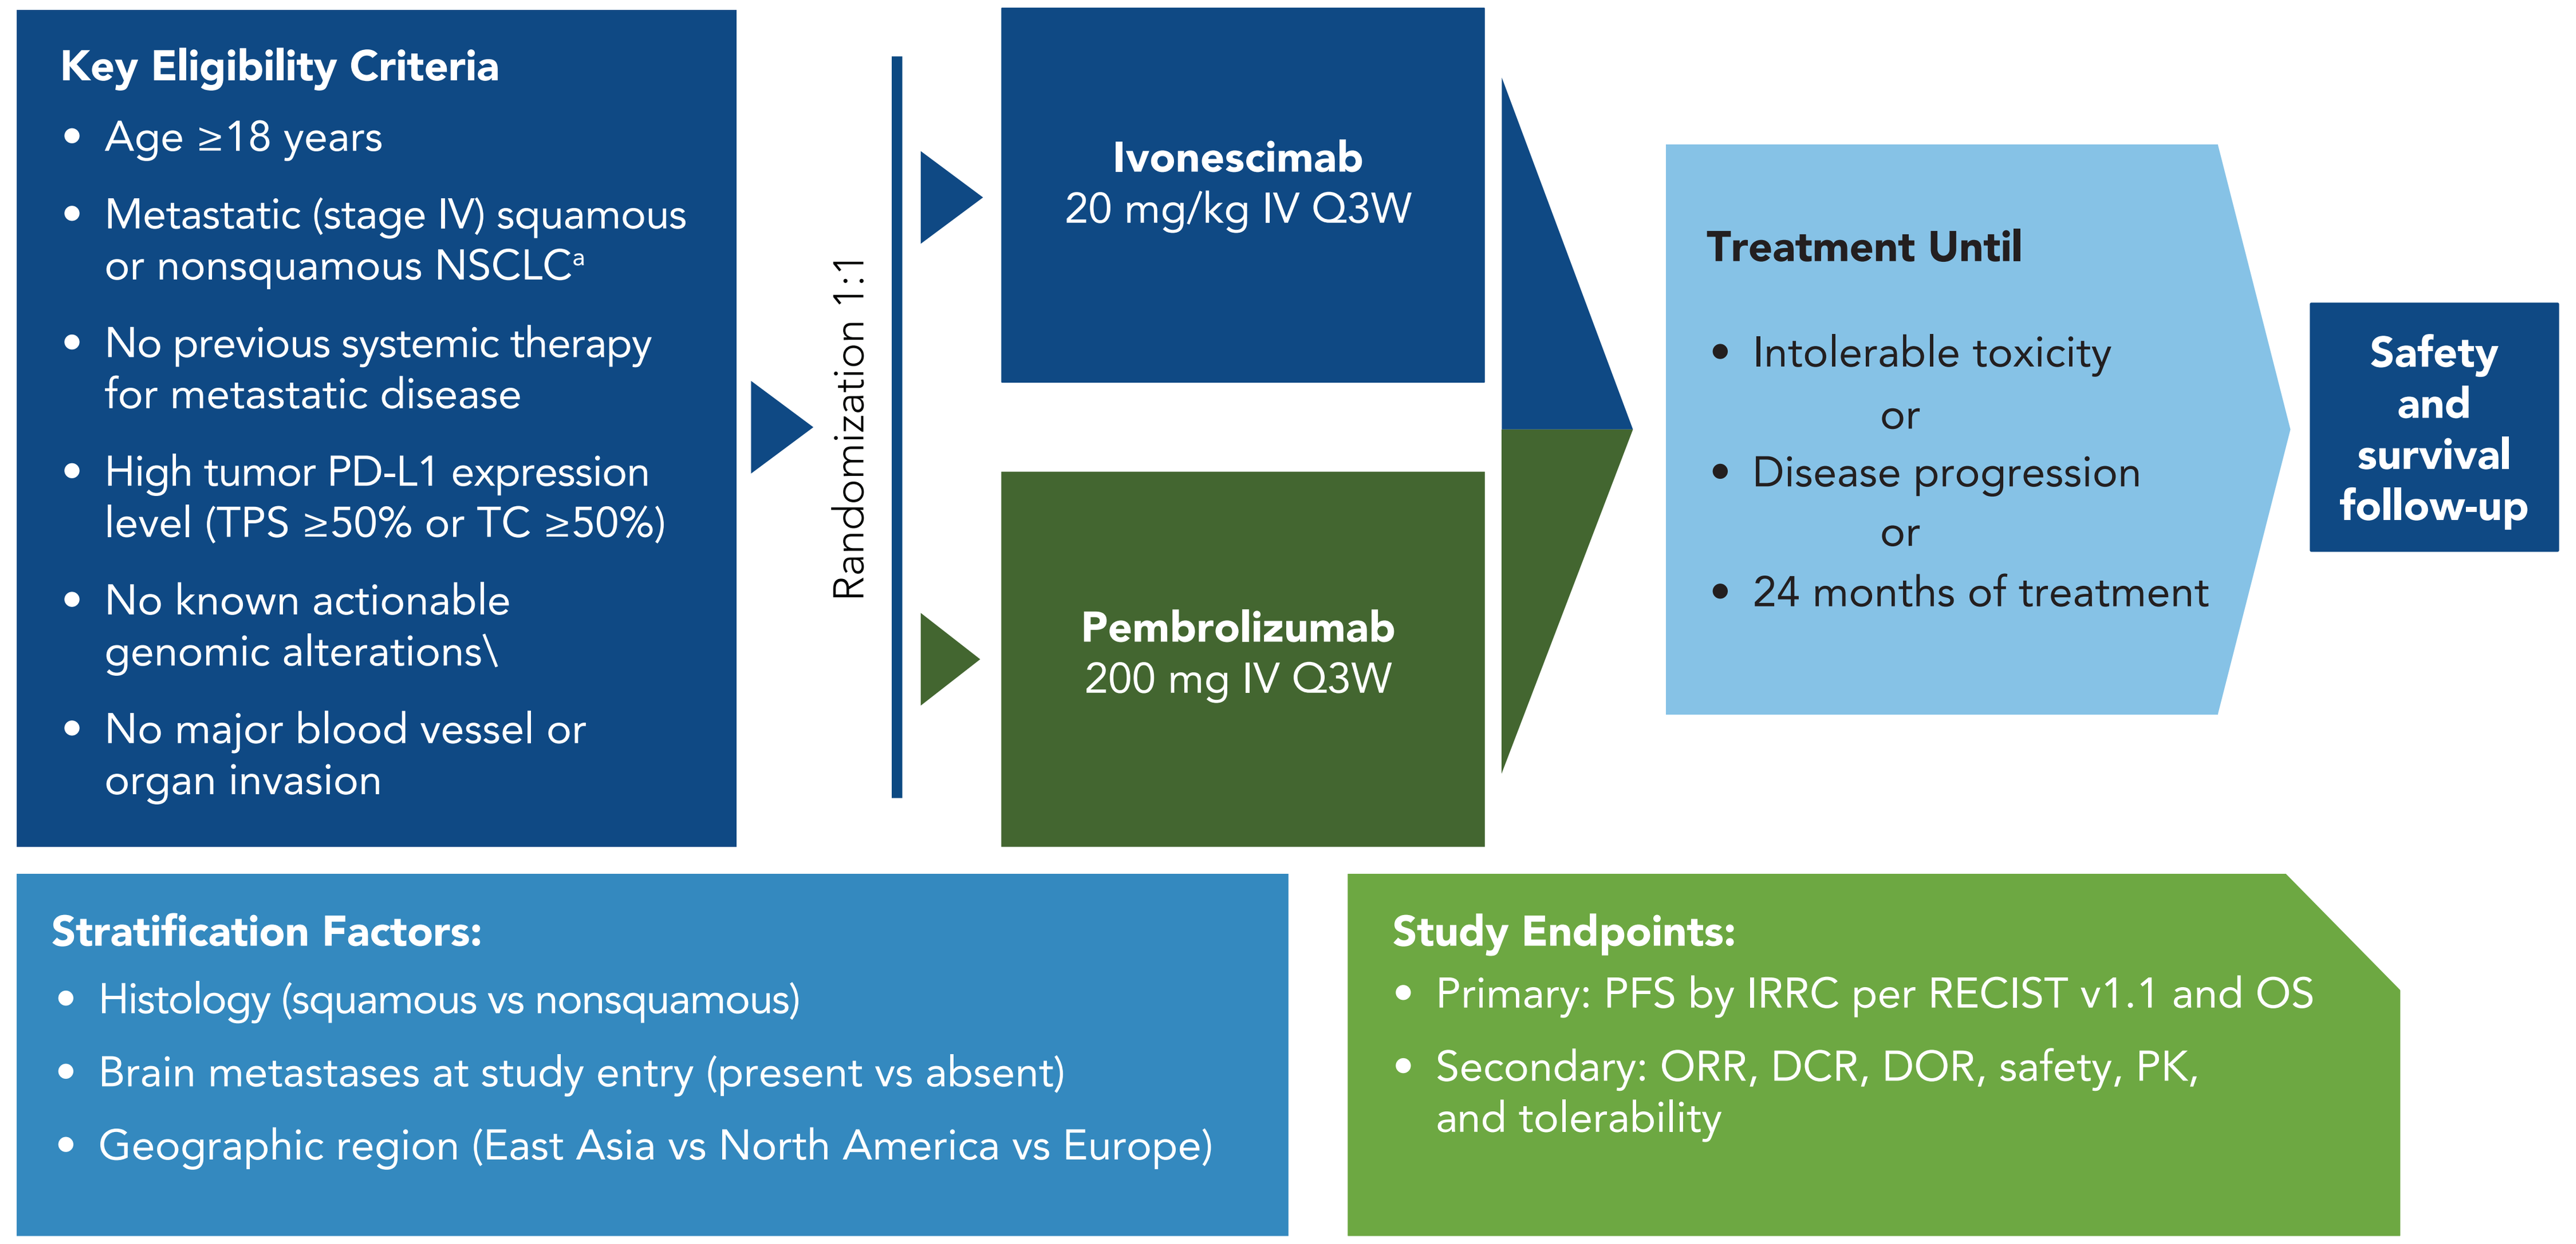
Key Eligibility Criteria
Age ≥18 years
Metastatic (stage IV) squamous or nonsquamous NSCLC[a]
No previous systemic therapy for metastatic disease
High tumor PD-L1 expression level (TPS ≥50% or TC ≥50%)
No known actionable genomic alterations\
No major blood vessel or organ invasion
Randomization 1:1
Ivonescimab
20 mg/kg IV Q3W
Pembrolizumab
200 mg IV Q3W
Treatment Until
Intolerable toxicity
or
Disease progression
or
24 months of treatment
Safety and survival follow-up
Stratification Factors:
Histology (squamous vs nonsquamous)
Brain metastases at study entry (present vs absent)
Geographic region (East Asia vs North America vs Europe)
Study Endpoints:
Primary: PFS by IRRC per RECIST v1.1 and OS
Secondary: ORR, DCR, DOR, safety, PK, and tolerability

AJCC, American Joint Committee on Cancer; DCR, disease control rate; DOR, duration of response; IRRC, independent radiology review committee; IV, intravenously; NSCLC, non–small cell lung cancer; ORR, objective response rate; OS, overall survival; PD-L1, programmed cell death ligand 1; PFS, progression-free survival; PK, pharmacokinetics; Q3W, every 3 weeks; RECIST v1.1, Response Evaluation Criteria in Solid Tumors, version 1.1; TC, percentage positive, tumor cells; TPS, tumor proportion score.

[a]According to the AJCC[5]; squamous or nonsquamous NSCLC subtype confirmed histologically or cytologically.

KEY ELIGIBILITY CRITERIA

Key Inclusion Criteria

- ≥18 years of age at time of enrollment
- mNSCLC (stage IV) per the American Joint Committee on Cancer (8th edition)[5]
- Eastern Cooperative Oncology Group performance status score of 0 or 1
- Histologically or cytologically confirmed squamous or nonsquamous NSCLC
- Tumor with high PD-L1 expression based on a 22C3 (TPS ≥50%) or SP263 (percentage positive tumor cells [TC] ≥50%) immunohistochemistry clinical assay, approved and cleared by local health authorities
- ≥1 measurable noncerebral lesion per Response Evaluation Criteria in Solid Tumors, version 1.1 (RECIST v1.1)
- No prior systemic treatment for mNSCLC

Key Exclusion Criteria

- Histologic or cytopathologic evidence of the presence of small cell lung carcinoma
- Known actionable genomic alterations (epidermal growth factor receptor [EGFR]; anaplastic lymphoma kinase [ALK]; proto-oncogene 1, receptor tyrosine kinase [ROS1]; or v-Raf murine sarcoma viral oncogene homolog B [BRAF] V600E) for which approved first-line therapies are indicated
- Any prior therapy for NSCLC in the metastatic setting
- Radiologic evidence of major blood vessel or organ invasion, or major blood vessel encasement with narrowing of the vessel or intratumor lung cavitation or necrosis that pose a significantly increased risk of bleeding per investigator
- Symptomatic central nervous system (CNS) metastases, CNS metastases with hemorrhagic features, CNS metastasis ≥1.5 cm, CNS radiation within 7 days before randomization, potential need for CNS radiation within the first cycle, or leptomeningeal disease
- Other previous malignant neoplasm (including previously treated NSCLC) unless the patient has undergone curative therapy with no evidence of recurrence of the disease for 3 years before randomization
- Active autoimmune or lung disease necessitating systemic therapy
- Preexisting peripheral neuropathy that is grade ≥2 by Common Terminology Criteria for Adverse Events, version 5
- Severe infection within 4 weeks before randomization
- Major surgical procedures or serious trauma within 4 weeks before randomization
- History of noninfectious pneumonia necessitating systemic corticosteroids or current interstitial lung disease

TRIAL ENDPOINTS

Primary Endpoints

- Overall survival
- PFS, assessed by independent radiology review committee (IRRC) based on RECIST v1.1

Secondary Endpoints

- Objective response rate, disease control rate, and duration of response, assessed by IRRC based on RECIST v1.1
- Safety, assessed by incidence and severity of adverse events and clinically significant abnormal laboratory test results
- Pharmacokinetics of ivonescimab and immunogenicity assessments, occurring throughout the study

TRIAL STATUS

- This study is expected to enroll in North America, Europe, and Japan, and has started enrollment in the United States (**Figure 2**)
- The latest information on this study can be found at ClinicalTrials.gov, reference number NCT06767514

Figure 2. Study Locations

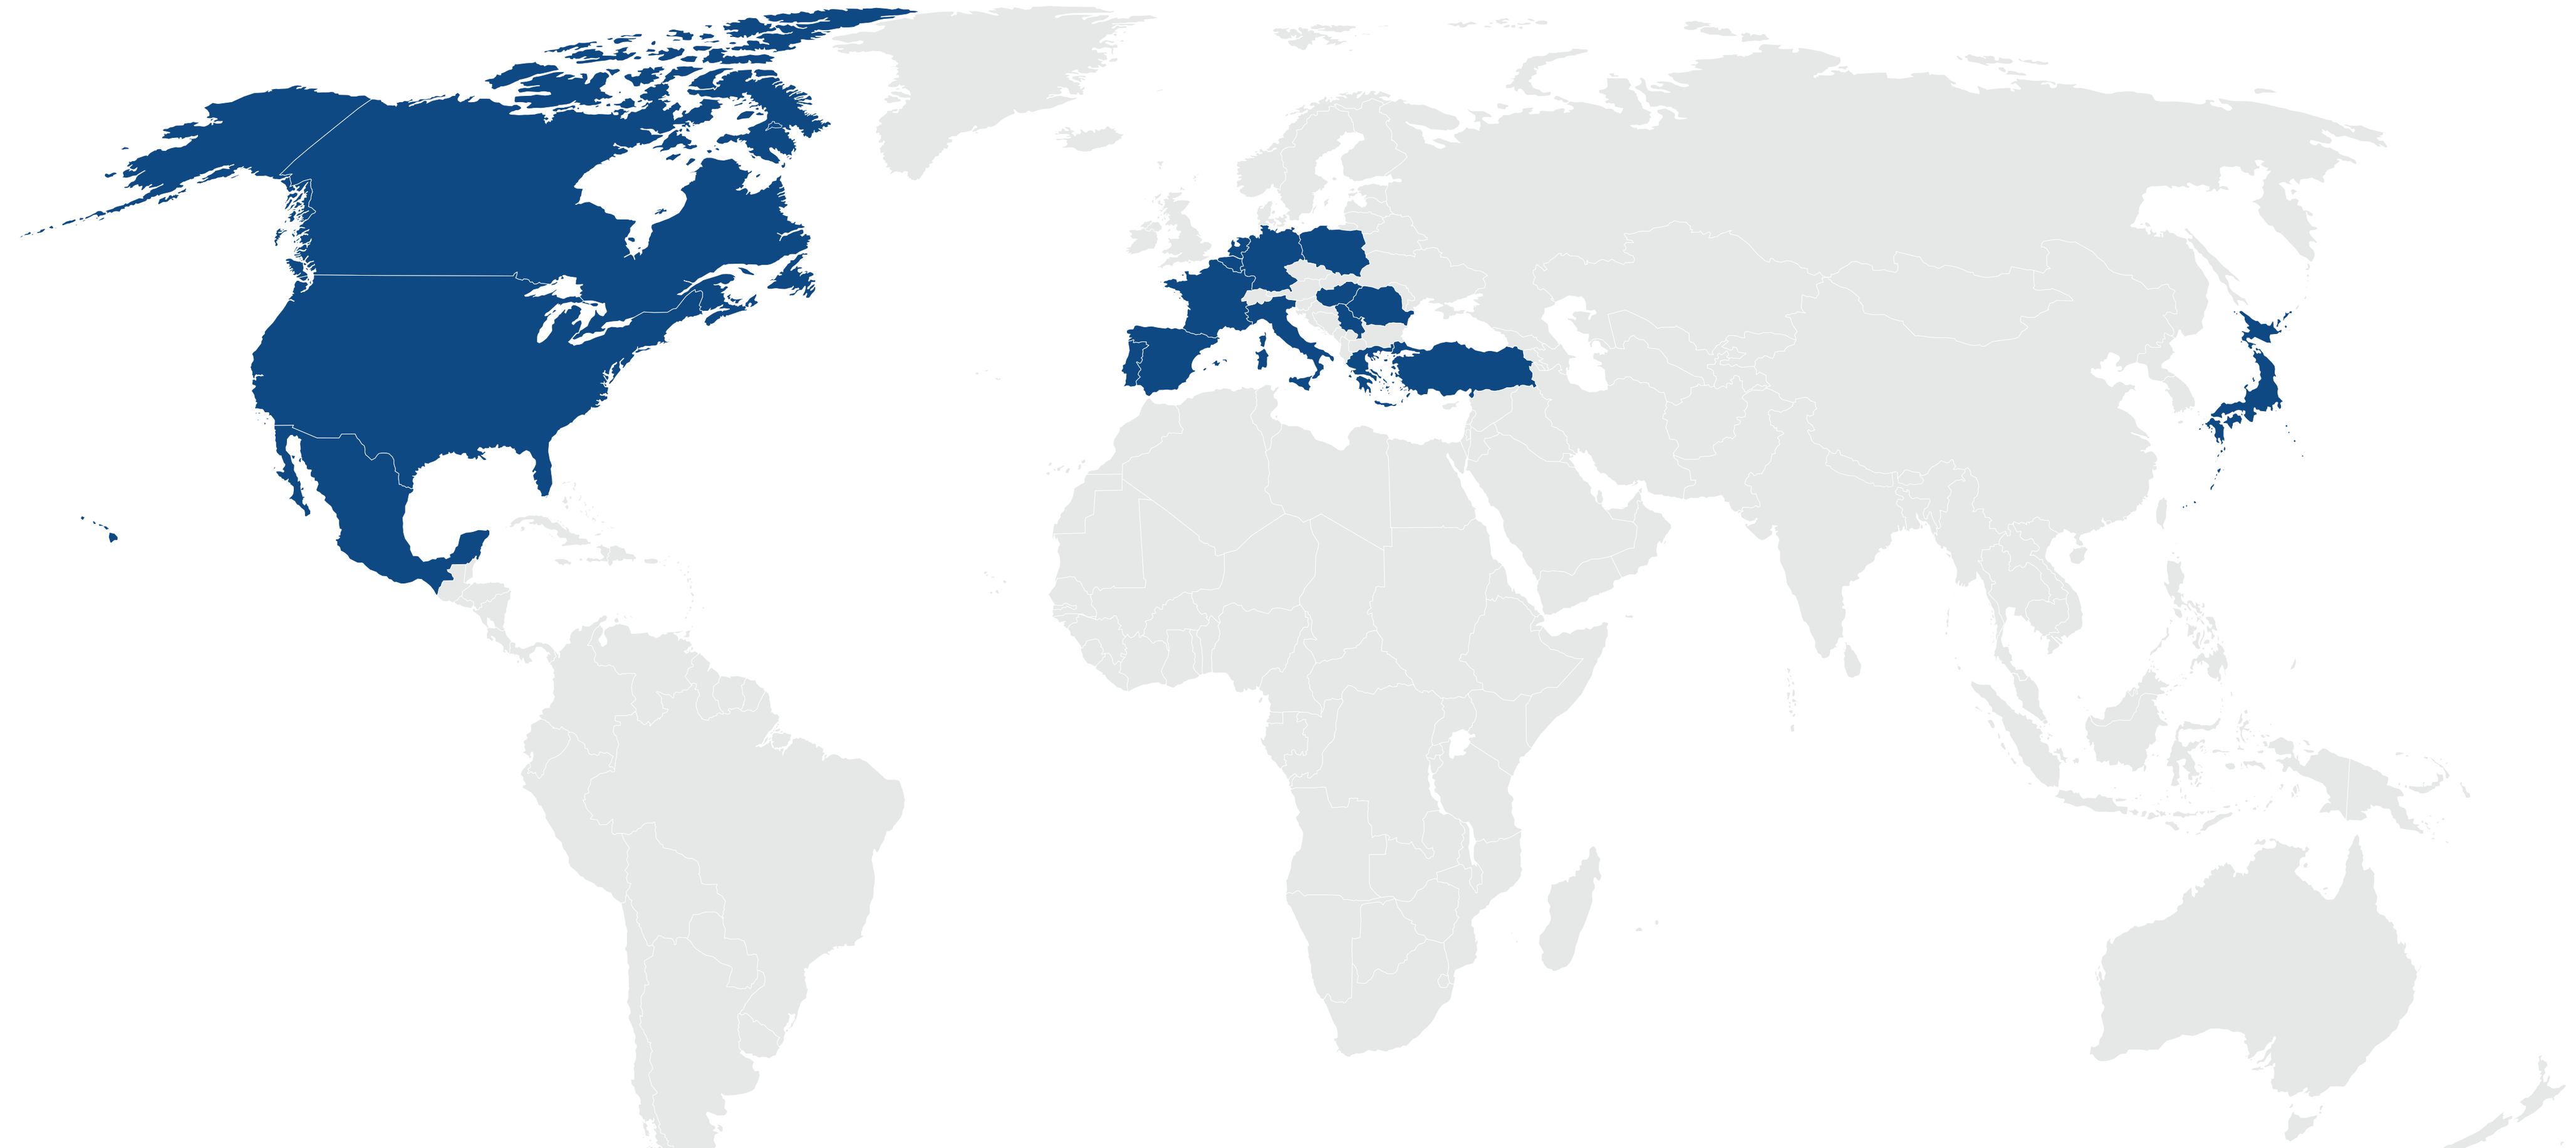

HARMONi-7 study site locations

ACKNOWLEDGMENTS

The authors thank the participants and their caregivers, the investigators, and all the site staff who have participated and who are participating in this study. This study is sponsored by Summit Therapeutics, Inc. Medical writing assistance was provided by MEDiSTRAVA (San Francisco, CA, USA) and was funded by Summit Therapeutics.

REFERENCES

1. Hendriks LE et al. *Ann Oncol.* 2023;34:358-376.
2. Socinski MA et al. *J Thorac Oncol.* 2021;16:1909-1924.
3. HARMONi-A study investigators. *JAMA.* 2024;332:561-570.
4. Xiong A et al. *Lancet.* 2025;405:839-849.
5. Admin MB et al. *AJCC Cancer Staging Manual.* 8th ed. New York: Springer; 2017.

DISCLOSURES

Financial Interests, Personal, Advisory Role: AstraZeneca, AbbVie, Arrivent, Bayer, Boehringer Ingelheim, Bristol Meyers Squibb, Daiichi Sankyo, Janssen, Johnson & Johnson, Gilead, GSK, Merck Sharp & Dohme, Novartis, Pfizer, Roche/Genentech, MundiPharma, Summit Therapeutics, Cullinan Oncology.

Ivonescimab is an investigational therapy not approved by any regulatory authority other than the National Medical Products Administration (NMPA) of China.

CONTACT INFORMATION

Please contact the presenting author Antonio Passaro at Antonio.Passaro@ieo.it for questions or comments.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

By use of the QR Code, you agree to the Summit privacy notice, located at smmttx.com.

Presented at the European Lung Cancer Congress (ELCC) 2025; March 26-29, 2025; Paris, France